Afirma Genomic Sequencing Classifier (GSC) and Xpression Atlas Molecular Findings in Bethesda III Thyroid Nodules:

Comparative Analysis of Nuclear Versus Other Atypia and Correlation with Histology

Xiaobing Jin,[1] Taylor B. Cavazos,[2] Mohammed Alshalalfa,[2] Joshua P. Klopper,[2] Laura Been,[3] Brian Smola,[1] Madelyn Lew,[1] Xin Jing[1] | 1. University of Michigan. 2. Veracyte, Inc. 3. Thyroid Cytopathology Partners

Disclosure: TBC, MA, and JPK are employees and equity owners of Veracyte, Inc. LB is the owner of TCP. All other authors have no disclosures.

UNIVERSITY OF MICHIGAN

veracyte

THYROID CYTOPATHOLOGY PARTNERS

BACKGROUND

- The 2023 Bethesda System for Reporting Thyroid Cytopathology (TBSRTC) divides atypia of undetermined significance (AUS) nodules into two major subcategories: nuclear atypia (AUS-N) and other (AUS-O).
- While studies have reported the malignant risk of AUS thyroid nodules differs according to the nature of atypia, little is known about the difference of molecular profiles between AUS-N and AUS-O thyroid nodules.
- This study aims to investigate the molecular findings between AUS-N and AUS-O thyroid nodules across a large, real-world cohort of fine needle aspiration (FNA) samples and to correlate these findings with histological outcomes.

DESIGN

- A retrospective study of consecutive thyroid FNA samples diagnosed as AUS and sent for Afirma Genomic Sequencing Classifier (GSC) testing.
- Afirma GSC classifier results, Xpression Atlas (expressed variants and fusions) results in GSC-suspicious samples, and tumor specific molecular signatures were analyzed and compared between AUS-N and AUS-O samples.
- A single center, retrospective analysis of histopathology specimens with AUS-N or AUS-O cytology were analyzed as well.

RESULTS

- Between April 2024 – December 2024, there were 5,345 paired cytology and Afirma GSC samples sent through Thyroid Cytopathology Partners (TCP), read as AUS, and stratified as AUS-N or AUS-O (Table 1).
 - AUS-N = 920
 - AUS-O = 4,425
- The Afirma GSC ensemble classifier reported GSC-(S)uspicious results in 58.2% of AUS-N, compared to 28.1% of AUS-O (p<0.01) (Table 2).
- Overall, there were more molecular variants in the AUS-N Afirma GSC-S nodules than in AUS –O (34.6% vs. 27.8%), mainly driven by the proportion of *BRAF*V600E. There was no difference in the overall proportion of fusions between the two groups though higher risk fusions (*RET, NTRK3, ALK*) were more common in the AUS-N group (Table 3).
- AUS-N were significantly more *BRAF*-like on *BRAF-RAS* score (BRS) and had higher ERK and follicular-mesenchymal transition (FMT) signature expression than AUS-O nodules (Figure 1).
- There were 201 surgically resected AUS, Afirma GSC-S nodules analyzed from the University of Michigan (Table 4).
 - There were significantly more malignancies in the AUS-N (28/82=34.1%) vs AUS-O (18/119=15.1%).
 - There were significantly more Papillary Thyroid Carcinomas (PTC) in the AUS-N (25/27=92.6%) compared to AUS-O (12/18=66.7%).

TABLE 1.
Demographics of the AUS cohort

Demographics		AUS-Nuclear (N=920)	AUS-Other (N=4,425)	P-value
Age	Mean [Q1, Q3]	57.02 [45.27, 69.27]	59.75 [49.32, 71.35]	<0.01
Nodule Size	Mean [Q1, Q3]	2.55 [1.55, 3.20]	2.67 [1.70, 3.30]	<0.01
Sex	Female	717 (77.9%)	3,429 (77.5%)	0.803
	Male	203 (22.1%)	996 (22.5%)	
Nodule Location	Isthmus	53 (5.8%)	222 (5.0%)	0.297
	Left	413 (44.9%)	1,901 (43.0%)	
	Right	454 (49.3%)	2,296 (51.9%)	
	Missing	0 (0.0%)	6 (0.1%)	

TABLE 2.
Afirma GSC ensemble classifier results by AUS category

Atypia	Total	GSC-B	GSC-S
Nuclear	920	385 (41.8%)	**535 (58.2%)**
Other	4,425	3,183 (71.9%)	**1,242 (28.1%)**

TABLE 3.
Expressed variants and fusions in Afirma GSC-S nodules by AUS categories

Total	Nuclear 535	Other 1,242	P-value
FUSIONS	**32 (5.9%)**	**55 (4.6%)**	**0.26**
PAX8::PAARG	9 (1.7%)	38 (3.1%)	0.11
RET	**5 (0.9%)**	**2 (0.2%)**	**0.029**
NTRK3	**7 (1.3%)**	**3 (0.2%)**	**0.011**
ALK	**4 (0.8%)**	**1 (<0.1%)**	**0.031**
BRAF	2 (0.4%)	2 (0.2%)	0.59
VARIANTS	**185 (34.6%)**	**345 (27.8%)**	**<0.01**
BRAFV600E	**65 (12.1%)**	**15 (1.2%)**	**<0.01**
BRAFK601	3 (0.6%)	19 (1.5%)	0.10
NRAS	51 (9.5%)	134 (10.8%)	0.45
HRAS	36 (6.7%)	98 (7.9%)	0.43
KRAS	7 (1.3%)	17 (1.4%)	1.0
DICER	3 (0.6%)	13 (1.1%)	0.42
TSHR	11 (2.1%)	15 (1.2%)	0.20
EIF1AX	1 (0.2%)	2 (0.2%)	1.0
OBSCN	0 (0.0%)	6 (0.5%)	0.19

FIGURE 1.
Prognostic molecular signatures by AUS category

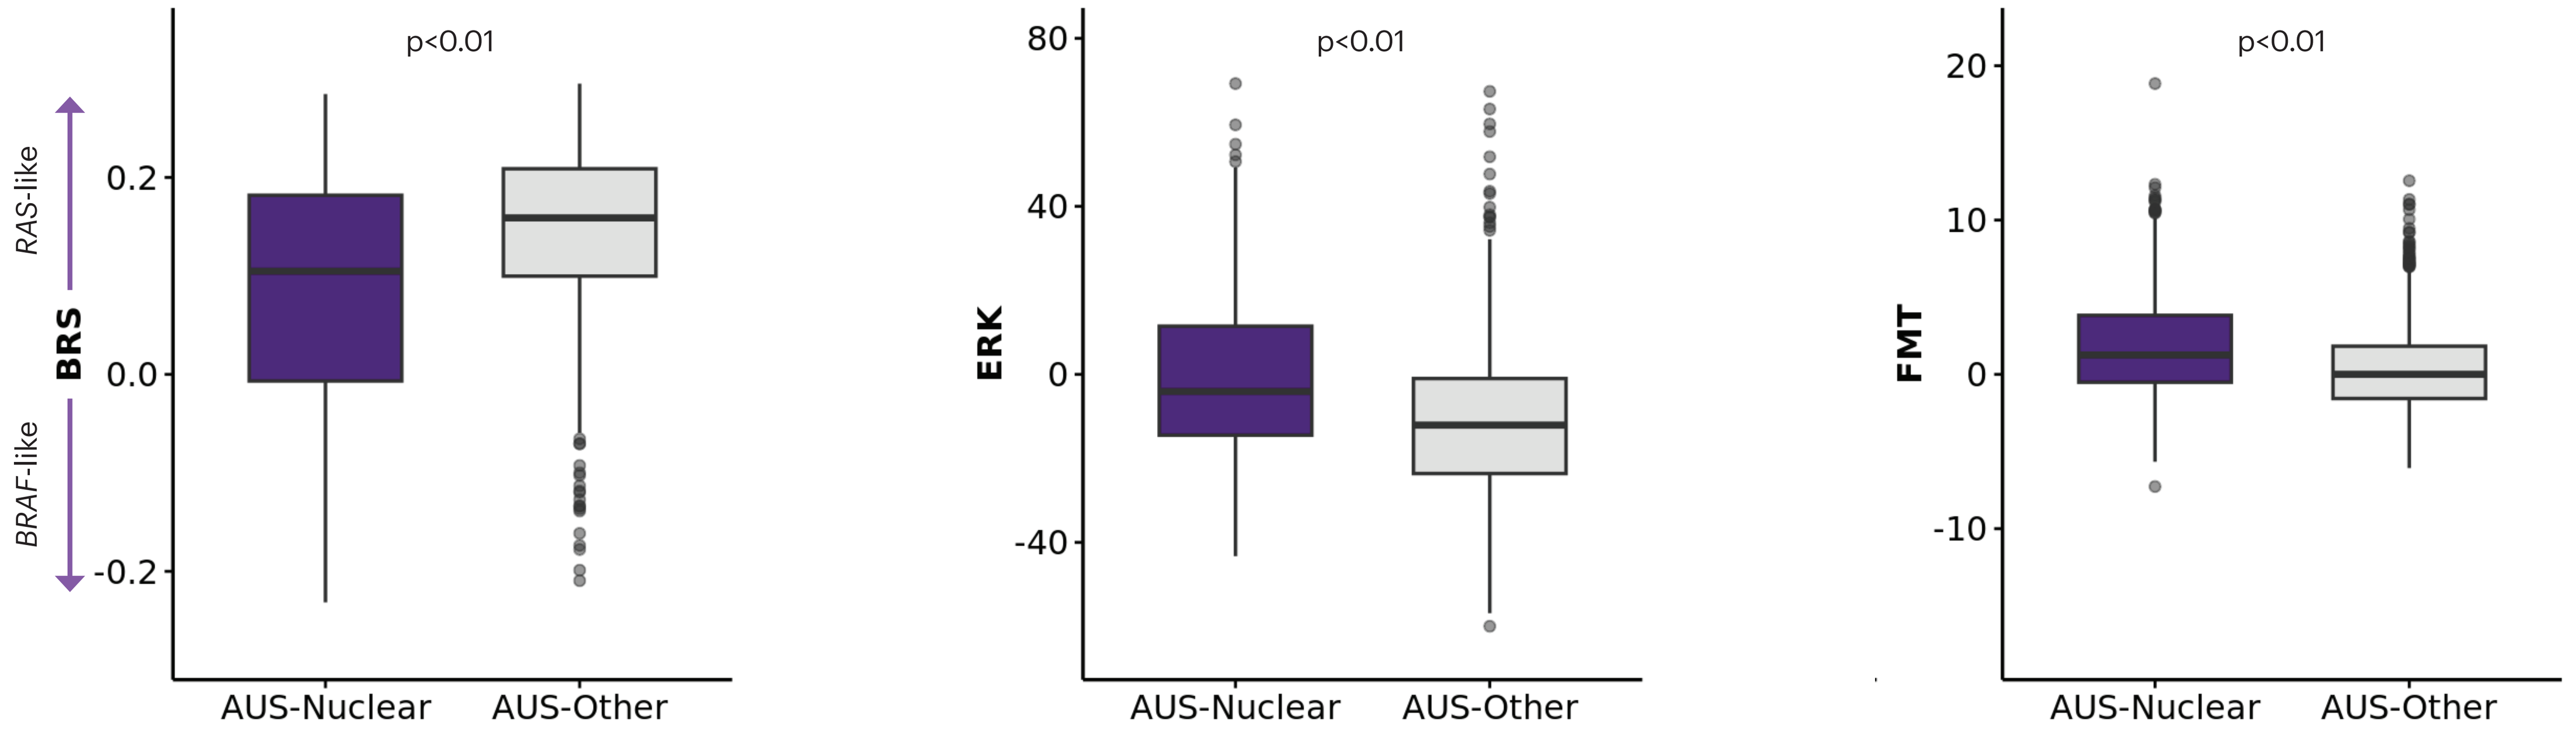

p<0.01
RAS-like
BRS
BRAF-like
0.2
0.0
-0.2
AUS-Nuclear
AUS-Other
ERK
80
40
0
-40
FMT
20
10
0
-10

BRS = *BRAF-RAS* score
FMT = follicular-mesenchymal transition

TABLE 4.
Histopathology Outcomes

A. Histopathology outcomes of AUS, GSC-S nodules

	AUS-N	AUS-O	P-value
Total	82	119	
Malignant Histopathology	28 (34%)	18 (15%)	
PTC	18 (22%)	10 (8.4%)	
PTC (follicular variant)	7 (8.5%)	2 (1.7%)	
Follicular carcinoma	1 (1.2%)	4 (3.4%)	
Oncocytic carcinoma	0	2 (1.7%)	
Medullary carcinoma	1 (1.2%)	0	
Lymphoma	1 (1.2%)	0	
Benign Histopathology	54 (66%)	101 (85%)	
Follicular nodular disease	30 (36.6%)	61 (51.2%)	
Follicular adenoma	16 (19.5%)	28 (23.5%)	
Oncocytic adenoma	2 (2.4%)	7 (6%)	
Lymphocytic thyroiditis	3 (3.7%)	2 (1.7%)	
NIFTP*	3 (3.7%)	3 (2.5%)	
Rate of Malignancy	28/82=34.1%	18/119=15.1%	**p=0.002**

*Noninvasive follicular thyroid neoplasm with papillary-like nuclear features

B. Comparison of Histopathology Outcomes Between AUS-N and AUS-O Thyroid Carcinomas

	AUS-N (n=27)	AUS-O (n=18)	P-value
Distribution of Carcinoma			
PTC	25 (92.6%)	12 (66.7%)	**p=0.02**
Non-PTC	2 (7.4%)	6 (33.3%)	
T Stage. No. (%)			
T1	17 (63%)	13 (72.2%)	p=0.42
T2	8 (29.6%)	3 (16.7%)	
T3	1 (3.7%)	2 (11.1%)	
Extrathyroidal Extension			
Absent	27 (100%)	18 (100%)	
Present	0	0	
Margin Status			
Positive	0	1 (5.6%)	p=0.4
Negative	27 (100%)	17 (94.4%)	
N Stage. No. (%)			
Nx, N0	25 (92.6%)	16 (88.9%)	p=0.08
N1a	0	2 (11.1%)	
N1b	2 (7.4%)	0	
M Stage. No. (%)			
Mx, M0	27 (100%)	18 (100%)	
M1	0	0	
Locoregional Recurrence			
Yes	0	0	
No	27 (100%)	18 (100%)	
Distant Metastasis			
Yes	0	1 (5.6%)	p=0.4
No	27 (100%)	17 (94.4%)	

CONCLUSIONS

- AUS-N have a higher frequency of Afirma GSC-S results than AUS-O thyroid nodules.
- AUS-N have a higher frequency of *BRAF*p.V600E mutations and higher risk fusions than AUS-O thyroid nodules.
- Molecular signature expression of ERK and FMT are higher in AUS-N than AUS-O thyroid nodules, suggesting the potential for more aggressive tumor behavior.
- Consistent with the molecular findings, amongst Afirma GSC-S samples, AUS-N thyroid nodules are more likely to represent malignancy, specifically PTC, than AUS-O thyroid nodules.
- These findings support AUS subclassification and molecular testing to enhance risk stratification and potentially guide personalized management of AUS thyroid nodules.

© 2026 Veracyte, Inc. All rights reserved. Veracyte, the Veracyte logo, and Afirma, are registered trademarks of Veracyte, Inc. and its subsidiaries in the US and selected countries. All other trademarks are the property of their respective owners in the United States and other countries. AG2007.1.2602